Determining Anti-cancer Efficacy of a Reversible LSD1 Inhibitor, EXS74539, in Primary AML Tissues with Limited Thrombocytopenic Effects

Rin Okumura, Manuela Lautizi, Norbert Hieger, Venu Thatikonda, Martin Senekowitsch, Tzi-Yang Lin, Giuseppe Fiume, Robert Sehlke, Thomas Winkler-Penz, Andrew Payne, Ross Paveley, Christophe Boudesco, Gregory Vladimer; *Exscientia*

Poster: 2289P

Copies of this poster obtained through QR (Quick Response) and/or text key codes are for personal use only and may not be reproduced without written permission of the authors.

INTRODUCTION

Acute myeloid leukaemia (AML) is characterised by distinct chromatin dynamics and aberrant histone methylation regulations. The reversible nature of histone methylation, controlled by histone methyl-transferases and demethylases, has become attractive for the design of new AML therapies. Among them, LSD1 has recently gained interest and tractability with the emergence of new LSD1 inhibitor (LSD1i) clinical candidates.

Here, we report the *ex vivo* activity of EXS74539 (‘539) on primary AML and healthy bone marrow (BM) samples.[1] ‘539 is a novel, potent and selective LSD1i, precision designed to combine a reversible mechanism of action with a preferable PK profile and CNS penetration.[2]

METHODS

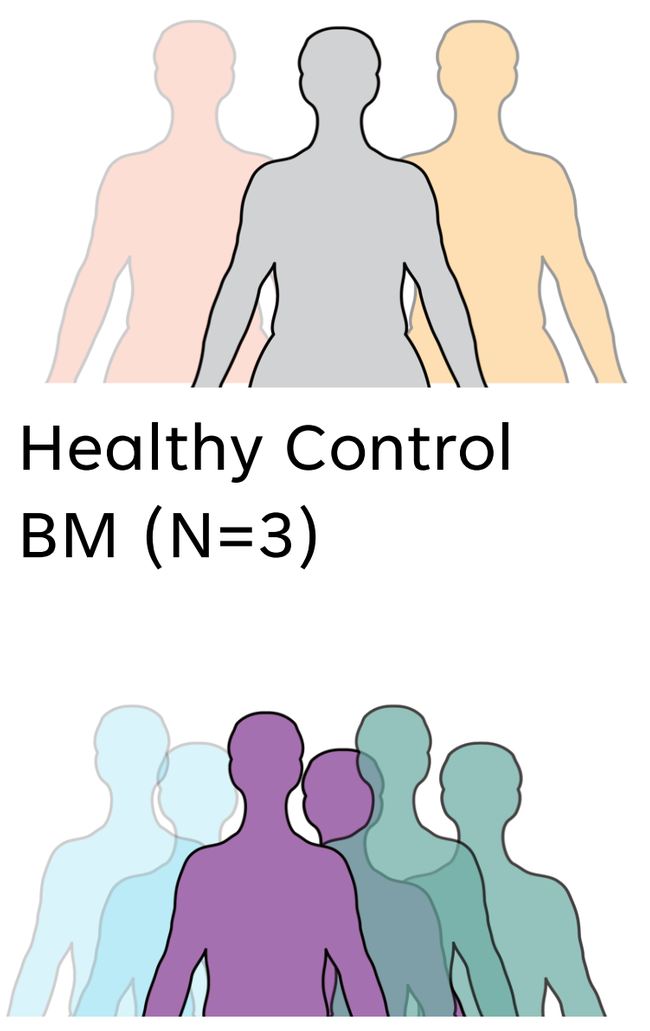

Healthy Control BM (N=3)

Primary AML BM (N=17)

Model systems: Primary newly diagnosed (ND) or relapsed/refractory (R/R) AML samples with characterised FAB subtypes were collected from partner centres under approved ethical committee reviewed protocols. Viably cryopreserved healthy BM were acquired from vendors. Samples underwent genomic / transcriptomic profiling and flow cytometry-based cellular composition estimation.

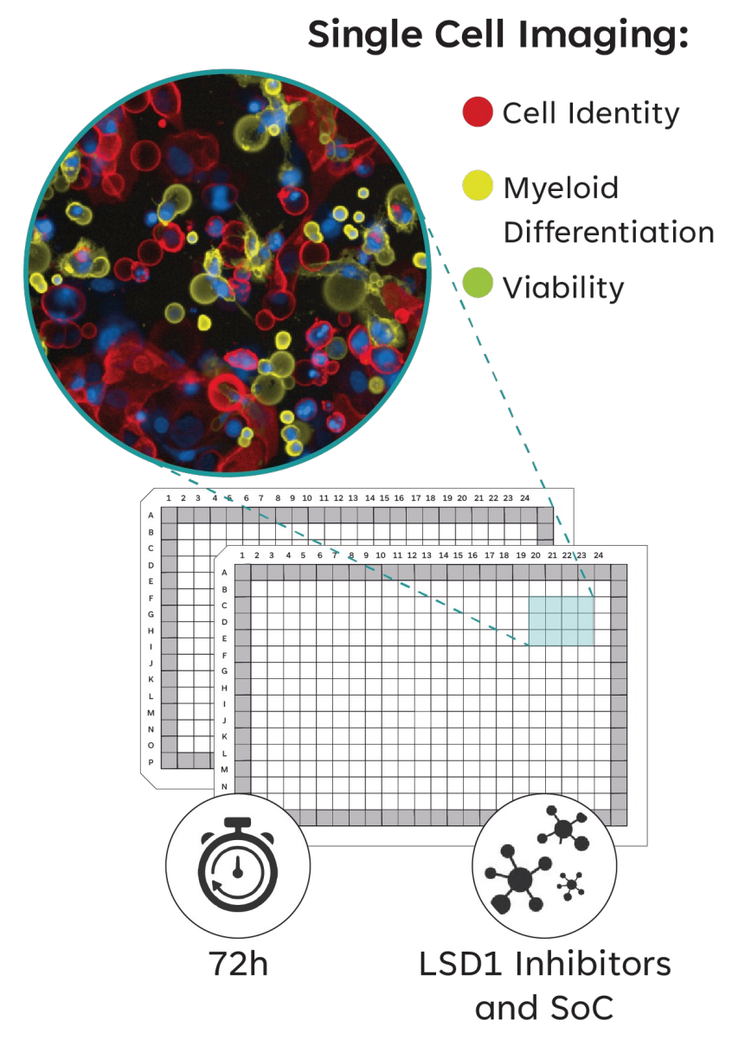

Single Cell Imaging:
Cell Identity
Myeloid Differentiation
Viability
72h
LSD1 Inhibitors and SoC

***Ex vivo* activity profiling:** Samples were exposed to standard of care (SoC), LSD1i alone or in combination for 72h *ex vivo* in culture media, and myeloid maturation marker expression / cell viability assessed using high content imaging platform. Data analysis is performed using our proprietary single cell image analysis algorithm.

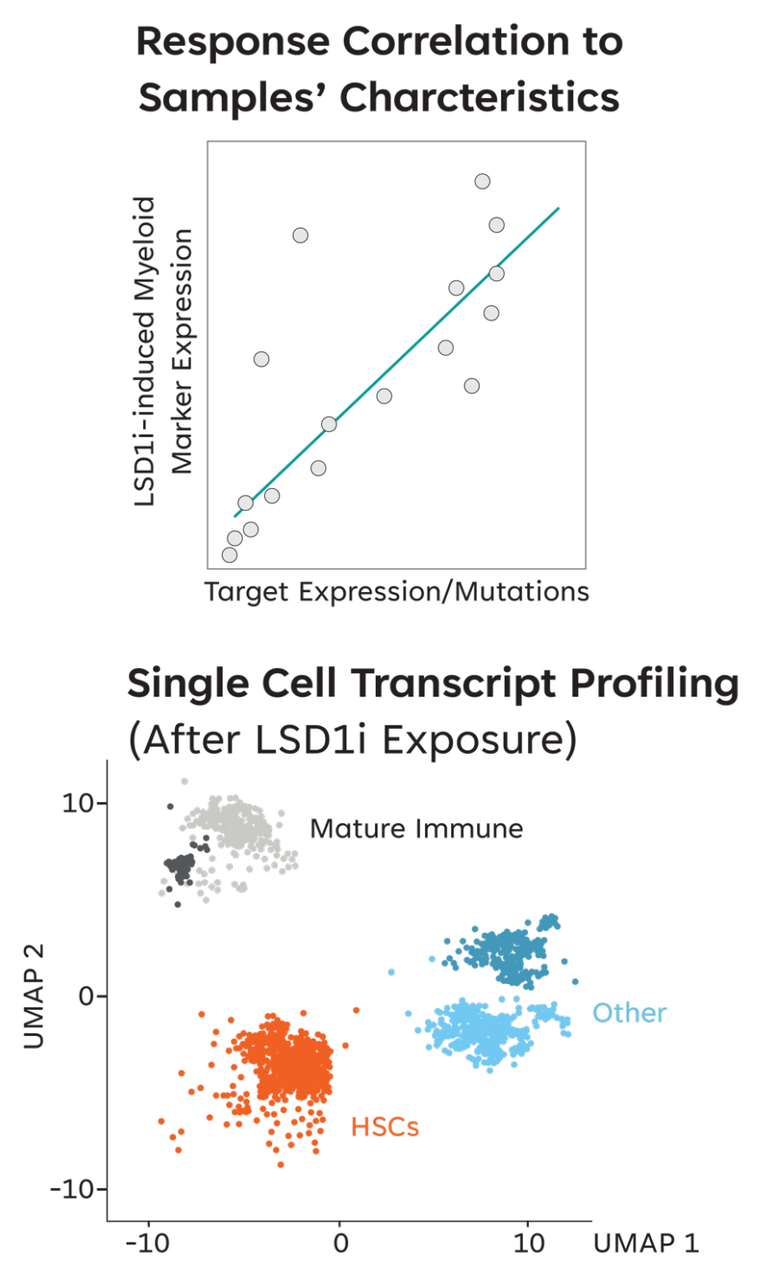

Response Correlation to Samples' Charcteristics
LSD1i-induced Myeloid Marker Expression
Target Expression/Mutations
Single Cell Transcript Profiling
(After LSD1i Exposure)
Mature Immune
Other
HSCs
UMAP 2
UMAP 1

Omics: RNA sequencing and single cell sequencing were performed at indicated timepoints and concentrations. Analyses were completed in R with Seurat package. Single cell data were corrected for cell cycle influence and batch effect, and cell populations predicted with SCINA tool, based on signature genes[5] and PanglaoDB[6].

RESULTS

Figure 1: LSD1i-induced *ex vivo* myeloid differentiation in primary AML cohort through measuring myeloid lineage marker expression

A) AML cohort (N=17)

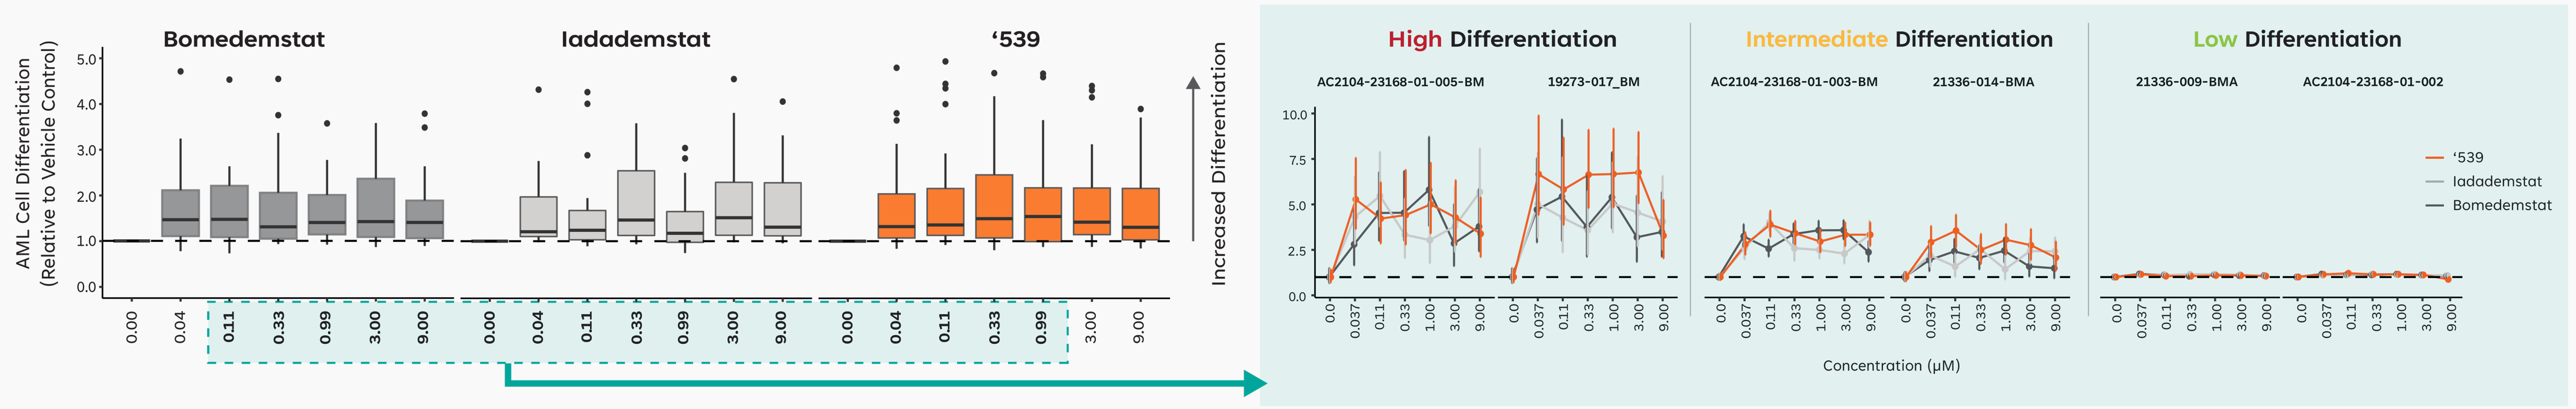

Bomedemstat
Iadademstat
‘539
AML Cell Differentiation (Relative to Vehicle Control)
Increased Differentiation
High Differentiation
Intermediate Differentiation
Low Differentiation
AC2104-23168-01-005-BM
19273-017_BM
AC2104-23168-01-003-BM
21336-014-BMA
21336-009-BMA
AC2104-23168-01-002
‘539
Iadademstat
Bomedemstat
Concentration (µM)

B) Healthy BM cohort (N=3)

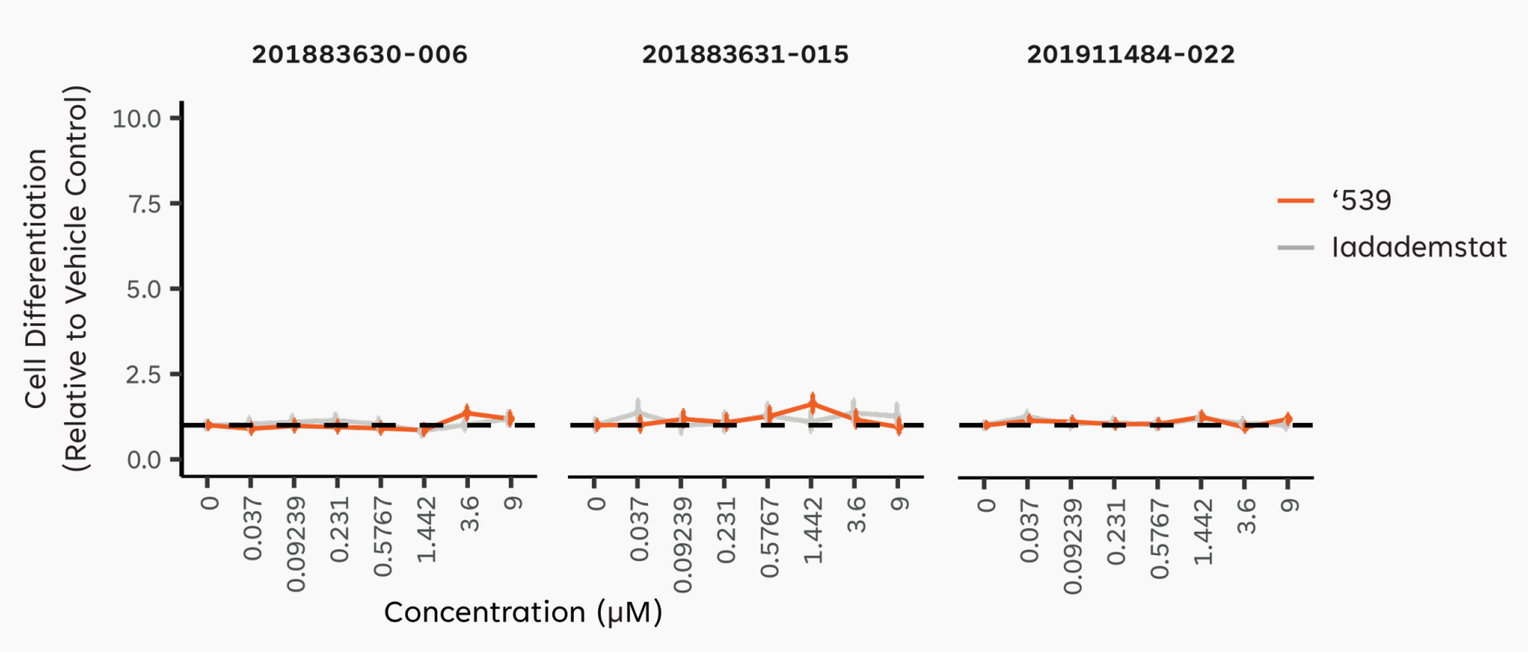

201883630-006
201883631-015
201911484-022
Cell Differentiation (Relative to Vehicle Control)
‘539
Iadademstat
Concentration (µM)

Figure 1: CD86 marker expression induction over 72h of LSD1 inhibition quantified using single cell phenotypic imaging. Y-axis: Fraction of $CD86^+$ expressing cells upon LSD1i exposure compared to vehicle control (DMSO) on **(A)** AML, **(B)** healthy BM samples. Similar results were obtained using another differentiation marker (CD11b).

Figure 2: Substantial variability of response magnitudes within the AML cohort

A) High patient-to-patient variability in LSD1i-induced myeloid cell differentiation

Patient ID	‘539	Bomedemstat	Iadademstat
73-017	5.123	3.012	3.442
04-01-005	3.308	2.607	3.018
36-014	2.934	1.430	1.496
04-01-003	2.361	2.102	1.757
384-65	2.075	1.985	2.115
73-014	1.680	1.320	1.280
73-003	1.277	0.874	0.936
36-010	1.069	0.935	0.933
37-001	1.028	1.346	1.026
04-01-001	0.988	0.906	0.836
04-01-004	0.620	-0.058	-0.073
36-019	0.409	0.386	0.406
37-018	0.268	0.084	0.138
04-01-002	0.121	0.143	0.140
37-003	0.114	0.111	0.049
36-002	0.078	0.021	0.004
36-009	0.039	0.012	0.029
630-006	0.010	N/A	0.040
3631-015	0.170	N/A	0.200
484-022	0.090	N/A	0.090

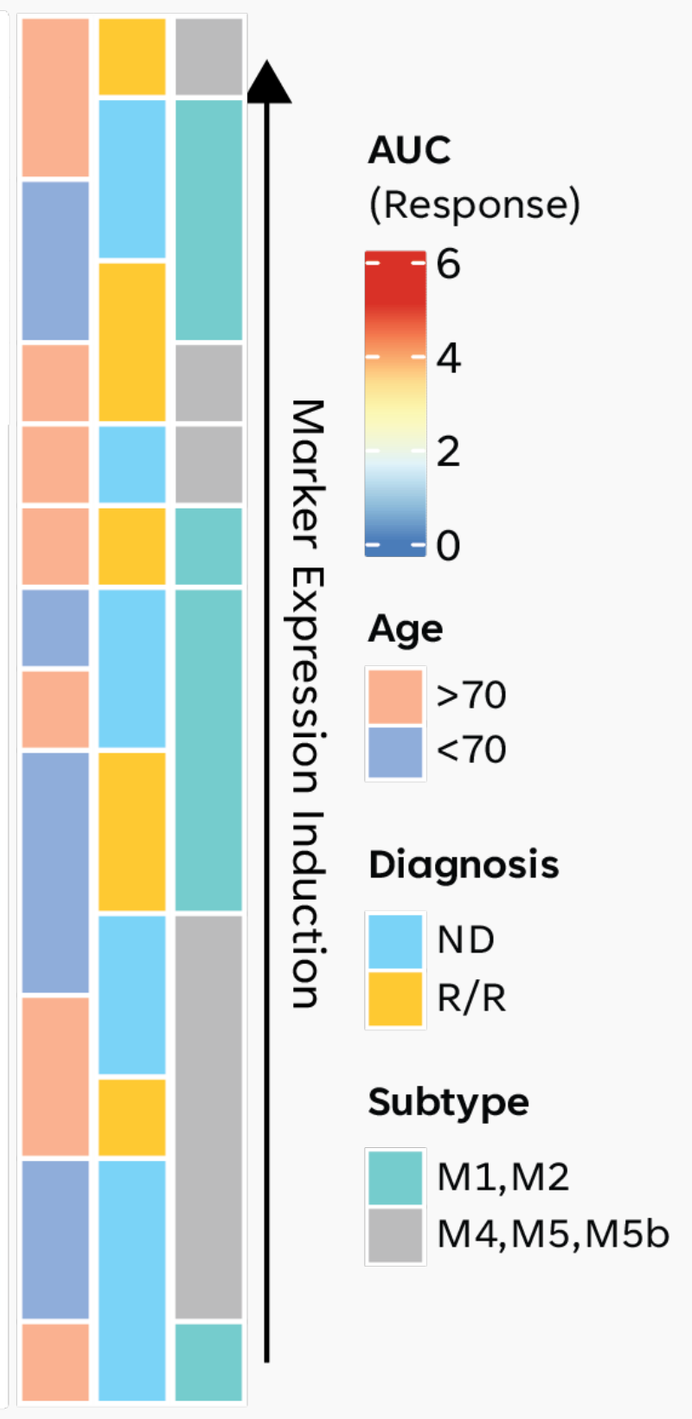

Metadata
Marker Expression Induction
Healthy BM
AUC (Response)
6
4
2
0
Age
>70
<70
Diagnosis
ND
R/R
Subtype
M1,M2
M4,M5,M5b

B) ‘539 differentiation profile is comparable to irreversible Phase 2 clinical candidate iadademstat

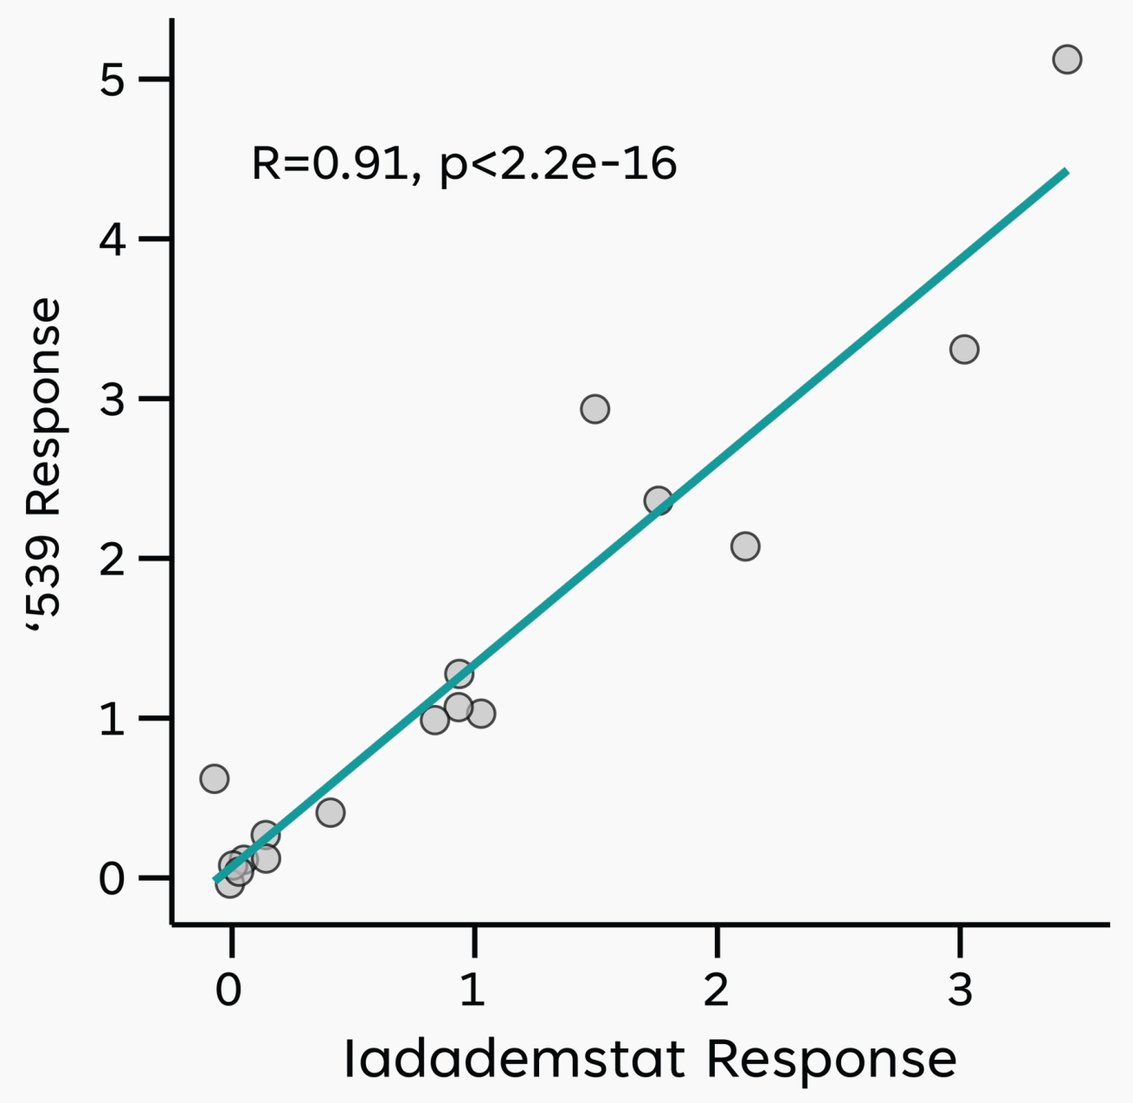

R=0.91, p<2.2e-16
‘539 Response
Iadademstat Response

Figure 2: CD86 marker expression post-LSD1 inhibition. **(A)** Heatmap represents the AUC of the **Figure 1A (right)/B** line charts, and for all patients. Demographics do not explain individual responses observed; ‘539 activity did not significantly correlate with treatment status, FAB disease subtype or age group (data not shown). **(B)** Spearman correlation plot of values represented in **Figure 2A**.

Figure 3: ‘539 synergises with first line SoC and targeted therapies

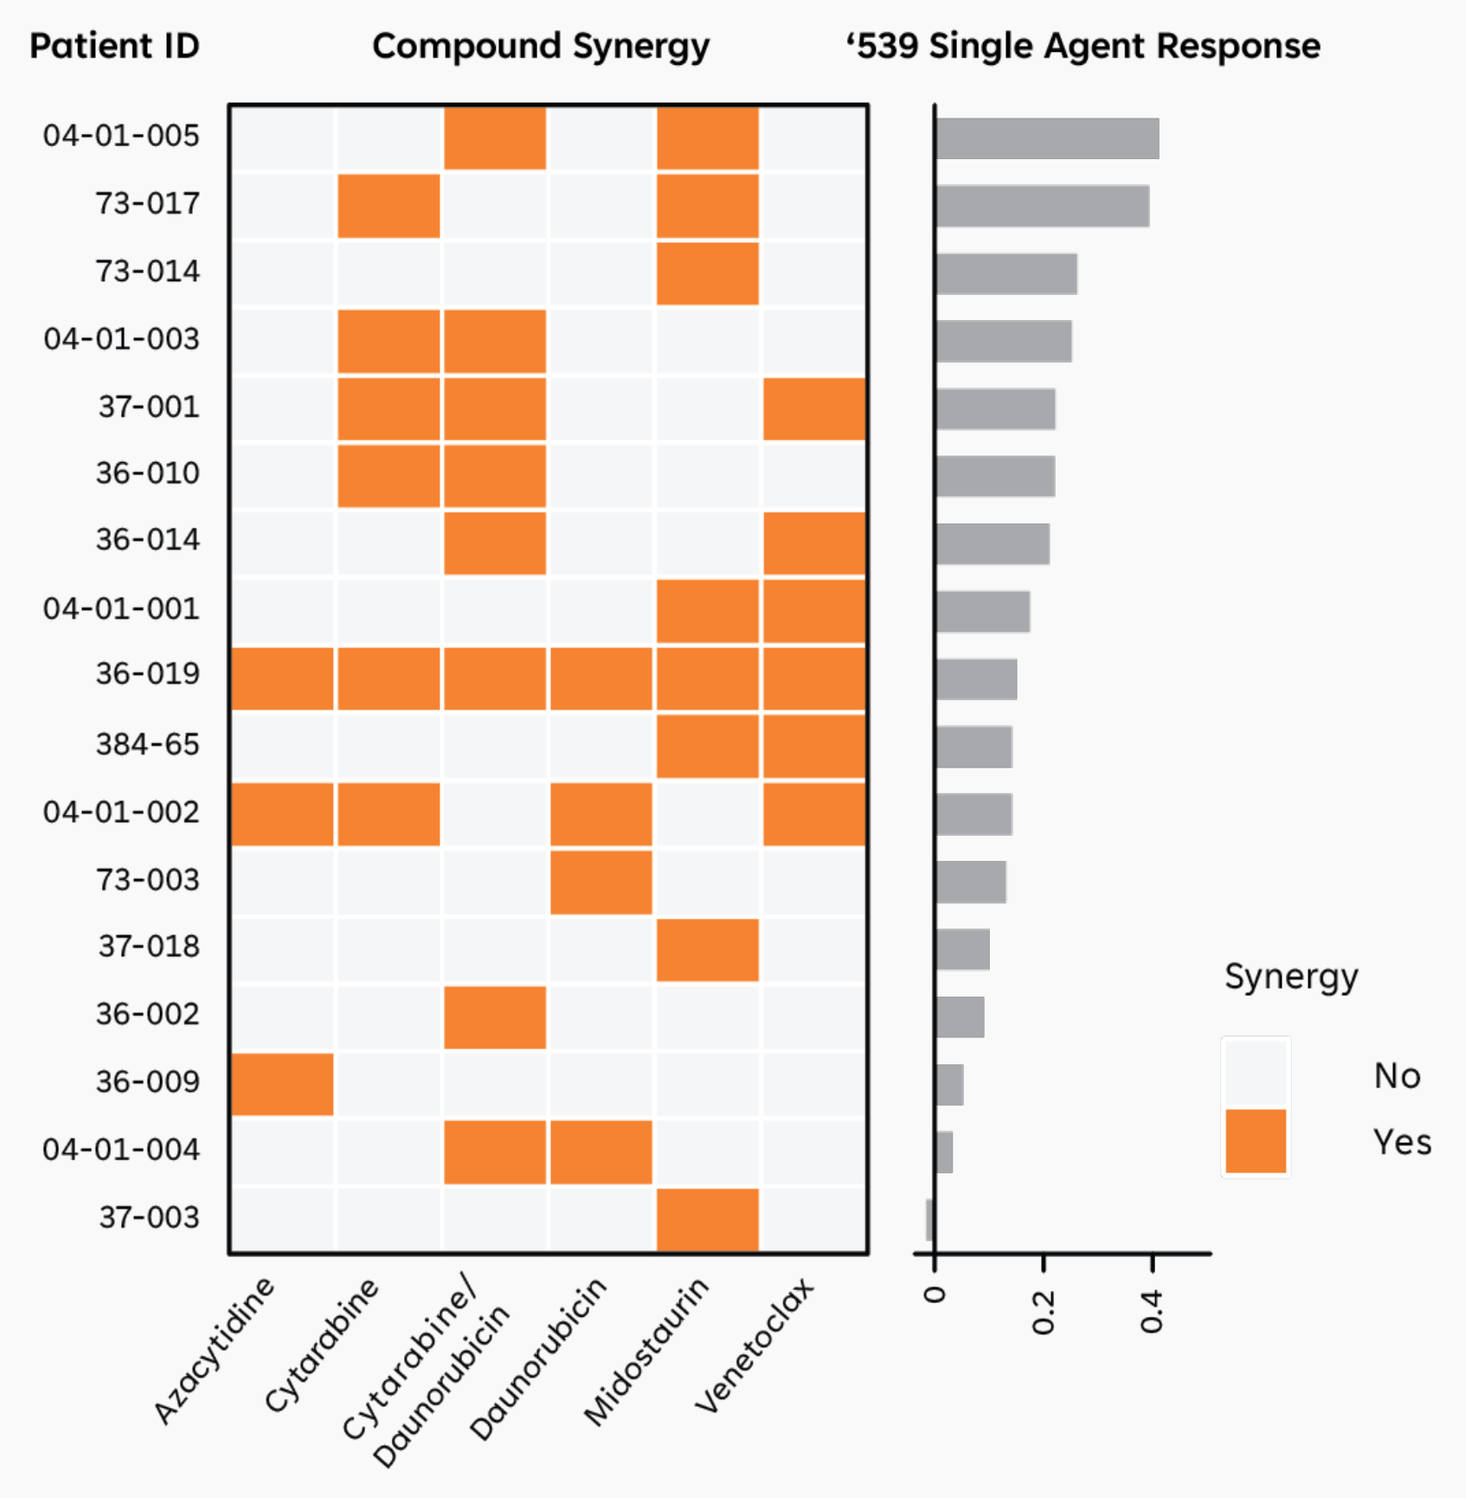

Patient ID
Compound Synergy
‘539 Single Agent Response
04-01-005
73-017
73-014
04-01-003
37-001
36-010
36-014
04-01-001
36-019
384-65
04-01-002
73-003
37-018
36-002
36-009
04-01-004
37-003
Azacytidine
Cytarabine
Cytarabine/ Daunorubicin
Daunorubicin
Midostaurin
Venetoclax
Synergy
No
Yes

Figure 3: Synergy potential between ‘539 and approved SoC and targeted therapies in primary AML samples. Combination potential established using HSA-index methodology. Significant synergy (orange boxes) indicates superior cancer cell induced loss of viability of the given drug combination compared to the respective single agent alone. Synergy observed across all diagnoses, age groups and subtypes.

Figure 4: ‘539 *ex vivo* response & baseline RNAseq association

A) No apparent correlation between ‘539 response and LSD1 expression

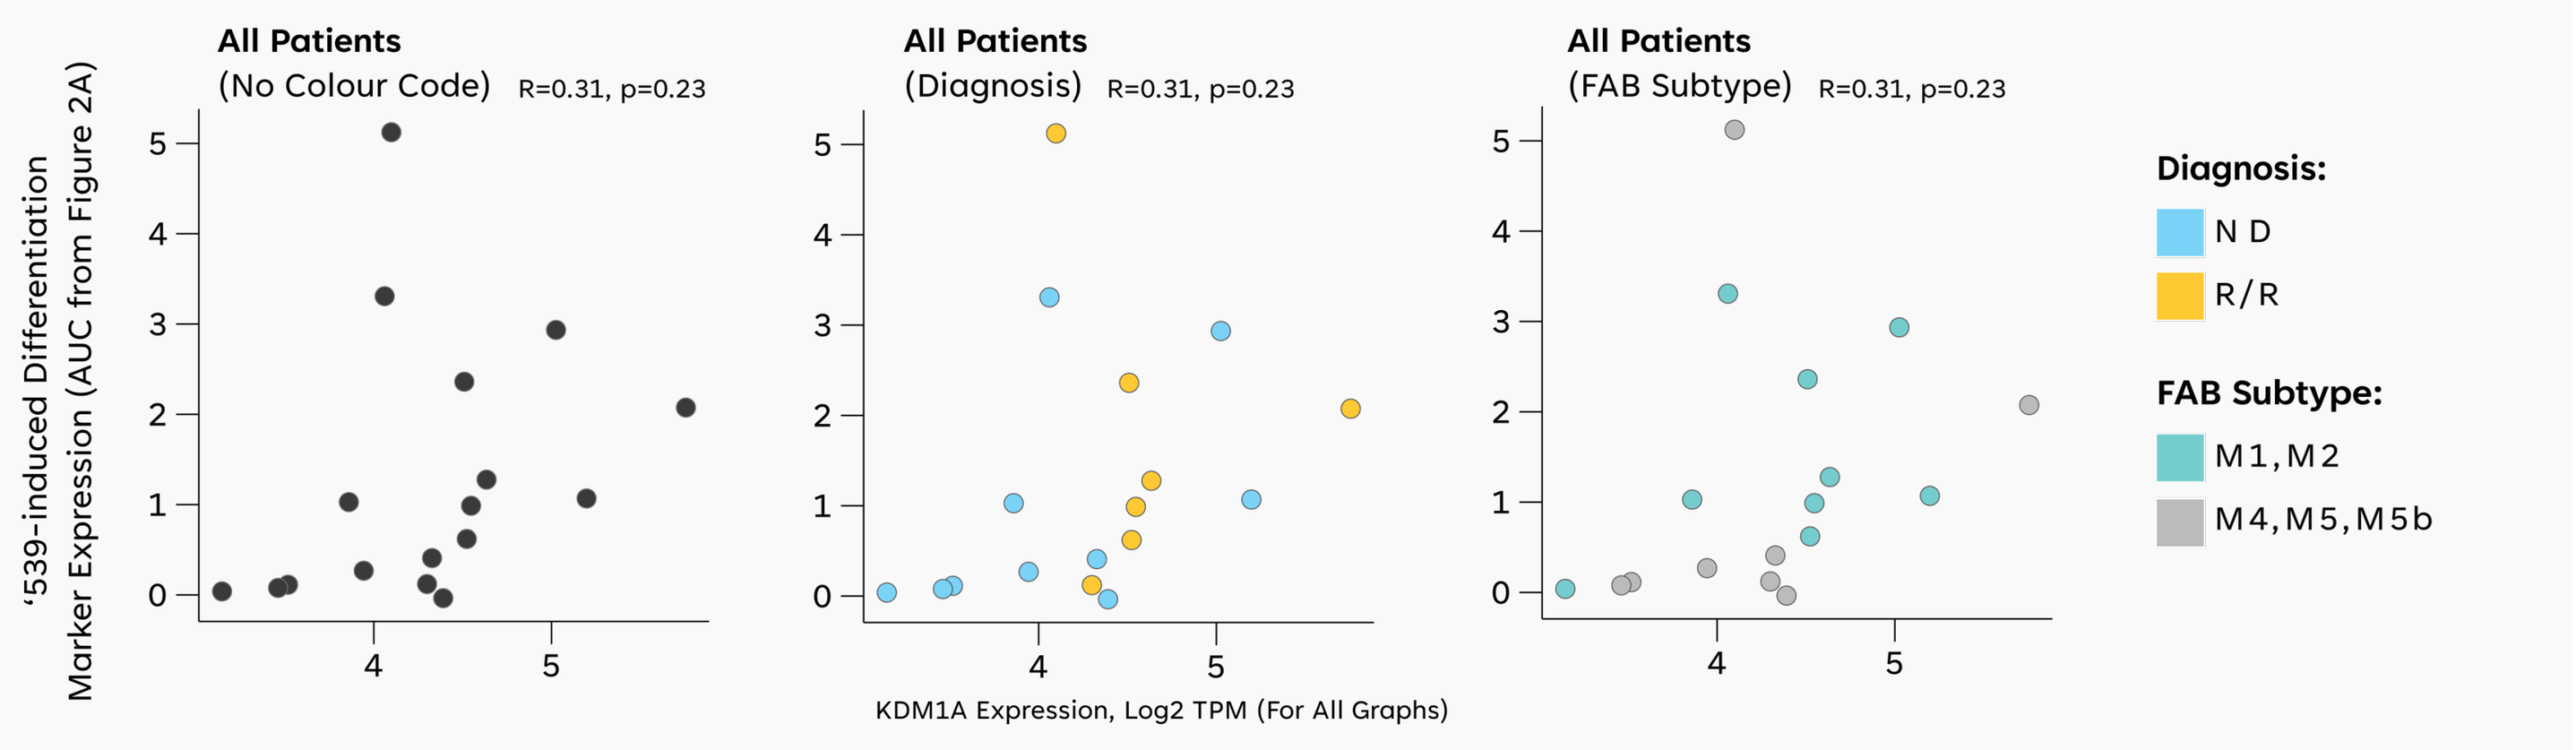

All Patients (No Colour Code) R=0.31, p=0.23
All Patients (Diagnosis) R=0.31, p=0.23
All Patients (FAB Subtype) R=0.31, p=0.23
‘539-induced Differentiation Marker Expression (AUC from Figure 2A)
KDM1A Expression, Log2 TPM (For All Graphs)
Diagnosis:
ND
R/R
FAB Subtype:
M1,M2
M4,M5,M5b

B) Trend for positive correlation between ‘539 response and LSC signature score

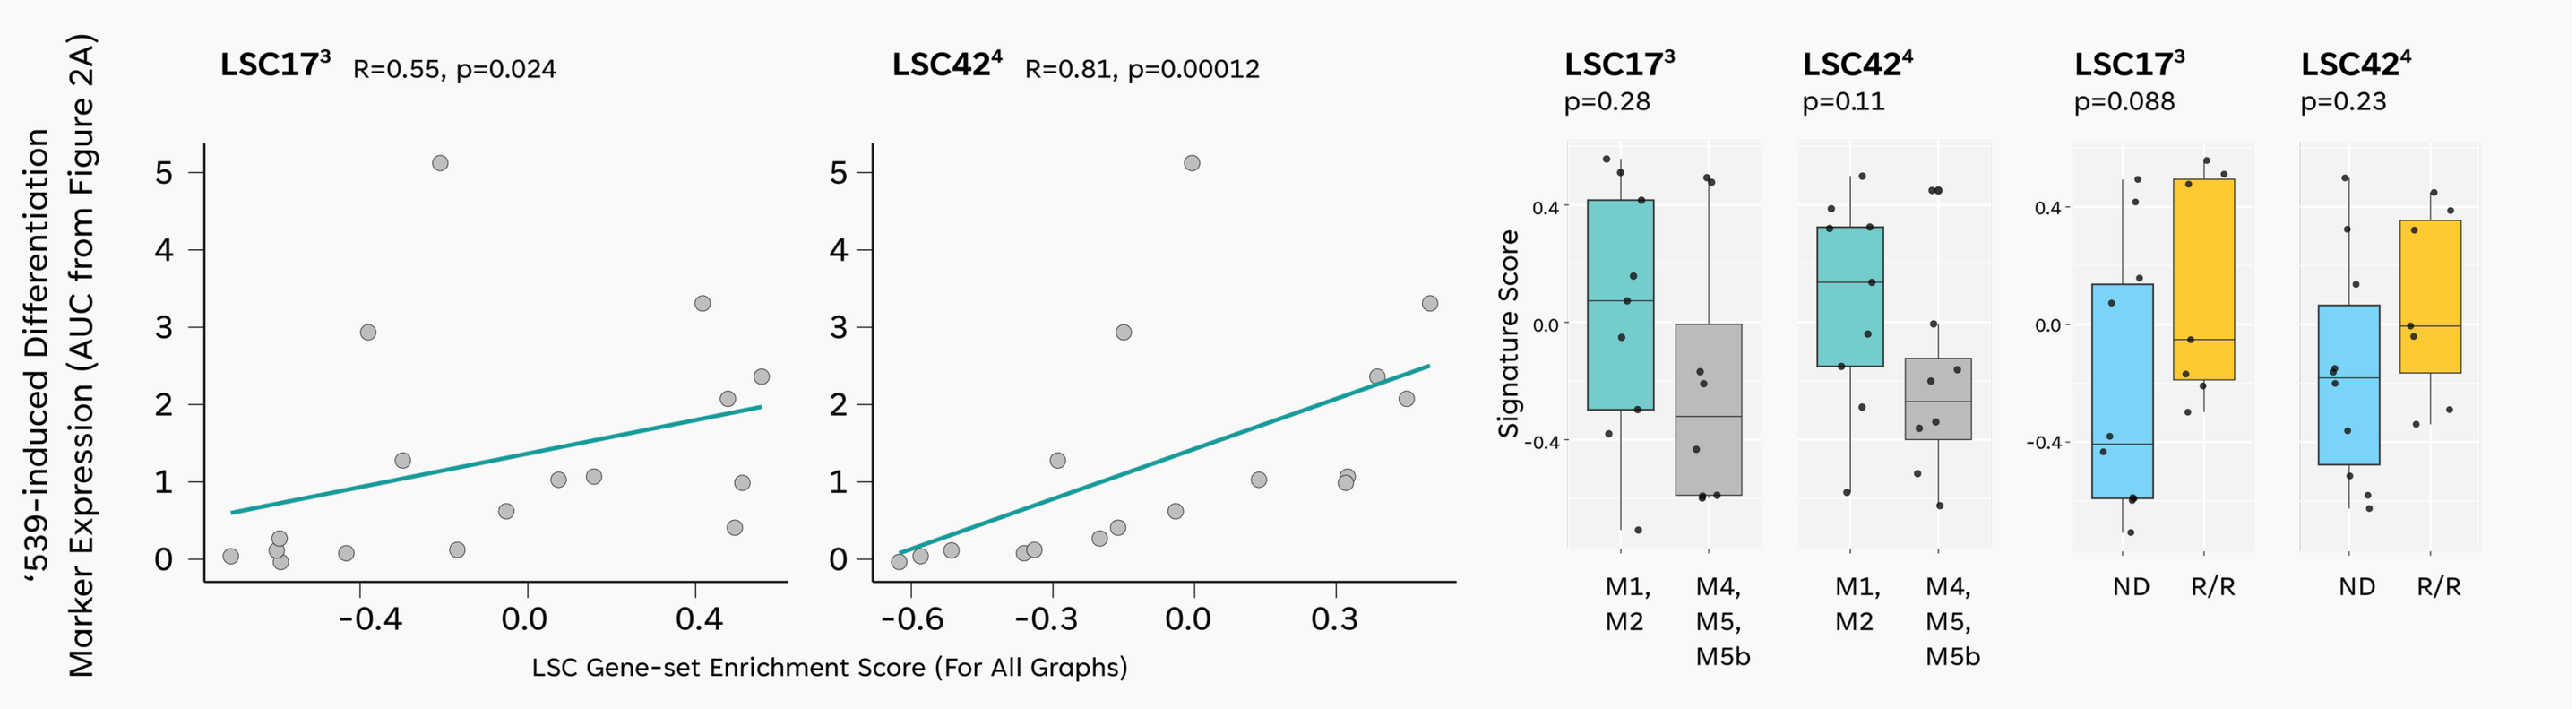

LSC17[3] R=0.55, p=0.024
LSC42[4] R=0.81, p=0.00012
‘539-induced Differentiation Marker Expression (AUC from Figure 2A)
LSC Gene-set Enrichment Score (For All Graphs)
LSC17[3] p=0.28
LSC42[4] p=0.11
LSC17[3] p=0.088
LSC42[4] p=0.23
Signature Score
M1, M2
M4, M5, M5b
ND
R/R

Figure 4: Spearman correlation of ‘539 induced marker expression induction in AML samples to the mRNA expression of LSD1 **(A)** or to the expression of two published leukaemic stem cell (LSC) signatures **(B, left plot)**. LSC signature as a function of the FAB subtype or treatment status is shown in **B, right plot**.

Figure 5: Single cell transcriptomics in primary AML uncovers leukaemic cell subtypes more likely modulated by ‘539

A) Primary AML sample cell types exposed to DMSO or LSD1i

B) LSD1 expression level per cell type

C) Cell proportions vary post '539 exposure

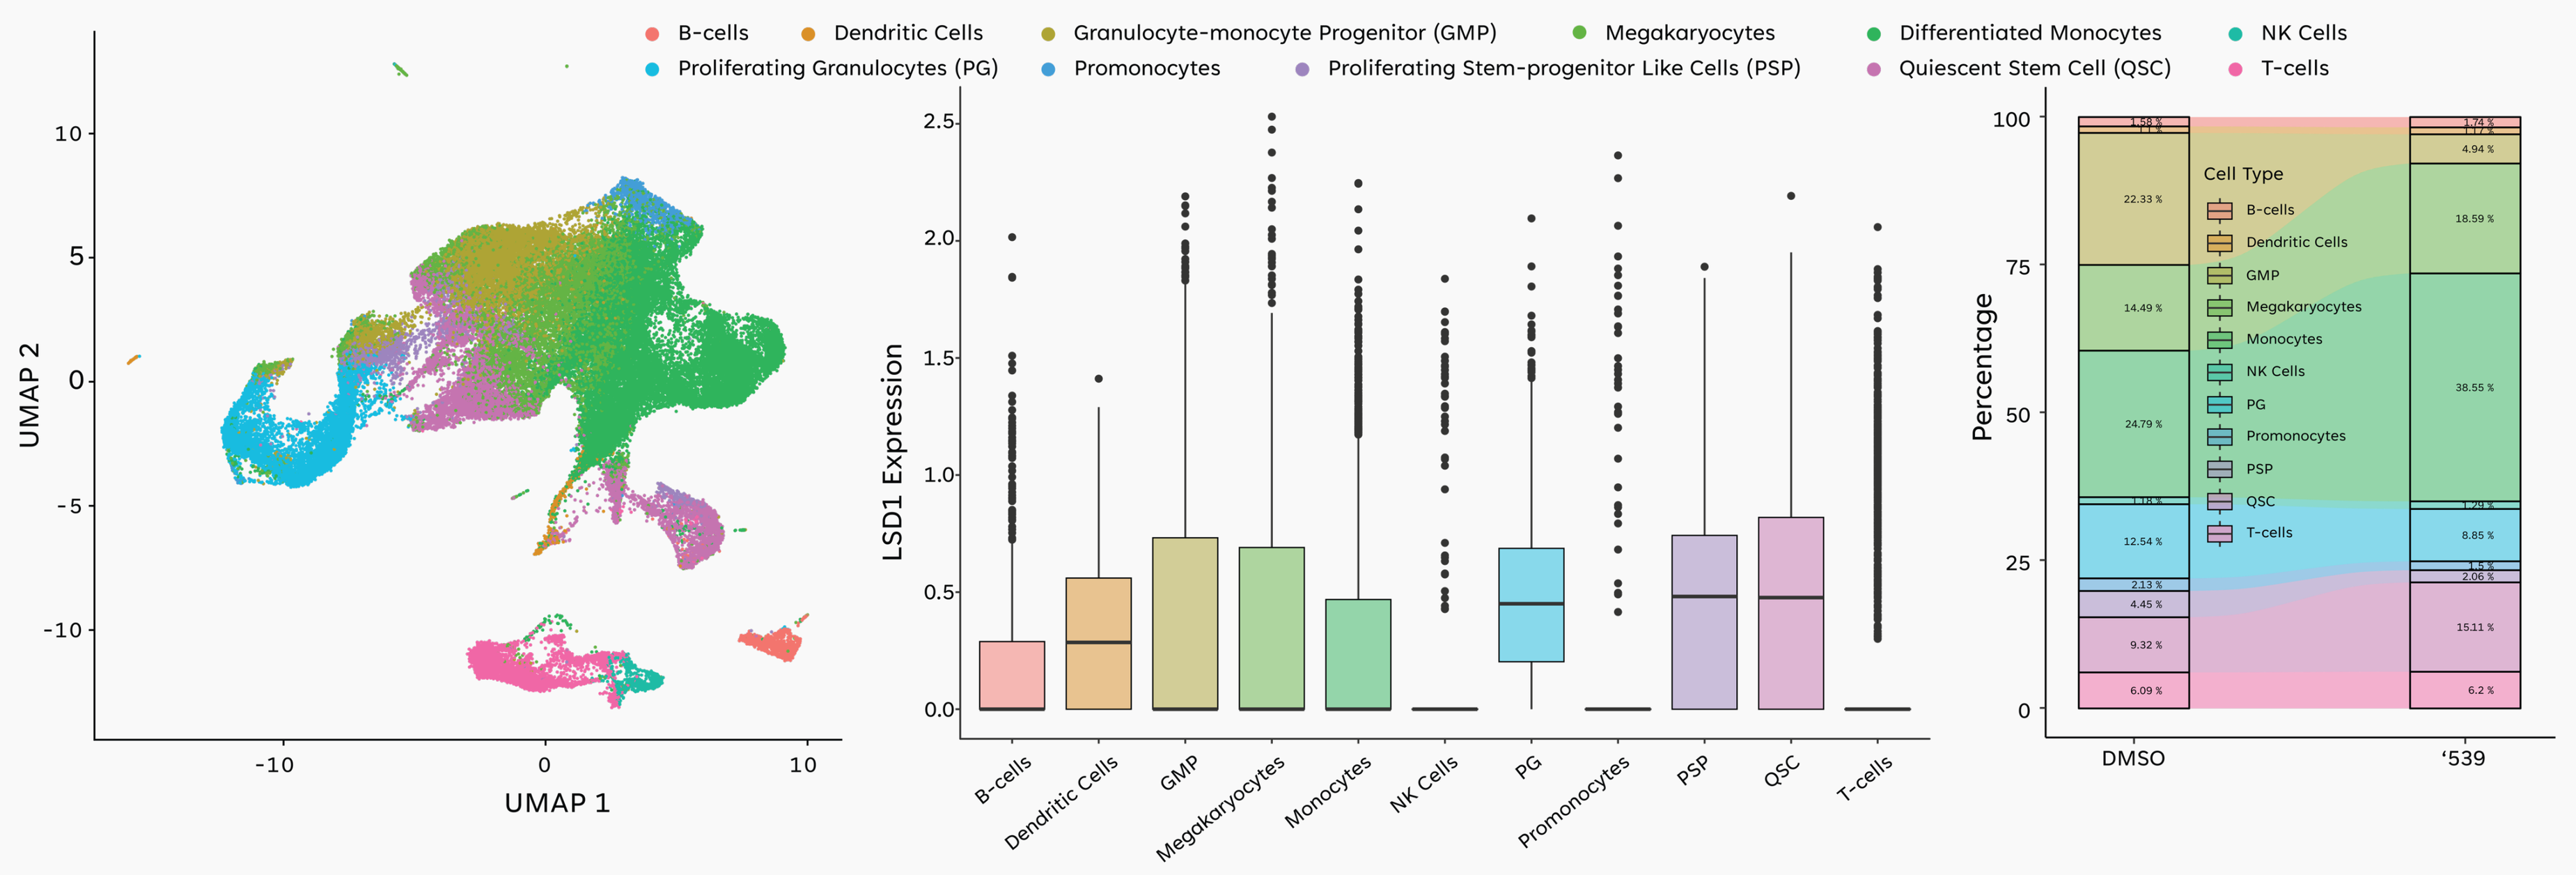

B-cells
Dendritic Cells
Granulocyte-monocyte Progenitor (GMP)
Megakaryocytes
Differentiated Monocytes
NK Cells
Proliferating Granulocytes (PG)
Promonocytes
Proliferating Stem-progenitor Like Cells (PSP)
Quiescent Stem Cell (QSC)
T-cells
UMAP 2
UMAP 1
LSD1 Expression
Percentage
Cell Type
DMSO
‘539

Figure 5: Single cell analysis of 6 AML patient samples upon 72h of ‘539 exposure. **(A)** UMAP visualisation of cell type identification. **(B)** LSD1 expression level per cell cluster (vehicle control treated). **(C)** Alluvial plot showing induced changes in cell proportions.

DISCUSSION

Our preclinical data demonstrate the *ex vivo* efficacy of ‘539 to induce AML cell differentiation after 72h incubation, while limiting healthy non-transformed bone marrow blast differentiation in the same conditions.

Exploring the response to ‘539 in AML, we observed a trend for positive correlation between ‘539-induced cell differentiation and sample stemness, defined by LSC signature score expression.

Leveraging our single cell omics capabilities further, we aim to detail ‘539-induced gene expression on AML cell subpopulations to generate patient enrichment hypotheses to identify patients more likely to respond to ‘539 in future clinical settings.

CONCLUSIONS

- Preclinically, ‘539 has potent *ex vivo* activity against primary human AML samples
- Our data supports the combination potential of ‘539 with first line clinical AML treatment strategies
- ‘539 has potential in a broad range of haematologic and oncologic diseases, including AML and small-cell lung cancer, and is currently progressing through IND/CTA-enabling studies

REFERENCES

1) Kornauth et al. Cancer Discov, 2022
2) Payne et al. Cancer Res, 2023
3) Stanley et al. Nature, 2016
4) Kolja et al. Nat Med, 2011
5) Zhang et al. Genes, 2019
6) Li et al. Leukemia, 2022
7) Franzén et al. Database, 2019

For more information: rokumura@exscientia.ai

Rin Okumura is an employee of Exscientia and owns stock and/or holds stock options in the Company.